Central Intake—A Streamlined Process for Diabetes Education Referrals to Improve Navigation of the System

Debbie E. Hollahan RN CDE, Sarah E. Christilaw, Elena M. Oreschina

Waterloo-Wellington Diabetes Regional Coordination Centre, Cambridge, Ontario

Aim

To create a streamlined process for referral to Diabetes Education Programs (DEPs)

Objectives

- To develop a central intake with one common physician referral form and a self-referral form
- To develop a model of care with clear definition of the roles of each diabetes program
- To identify triage criteria to improve access to the appropriate care
- To develop standard wait-times for education
- To provide timely access to information regarding the status of referrals (pending, booked, complete, reported) for primary care providers and patients
- To monitor wait-times of programs
- To help build and maintain capacity of diabetes education programs
- To standardize data collection in order to improve quality, monitor outcomes and implement appropriate changes

Methodology

This project combines qualitative and quantitative methods and is being conducted in four phases.

Central Intake Process Definition

The Centralized Intake (CI) Process is defined as beginning when a provider (physician) faxes the referrals to the central intake line, located within the Regional Coordination Centre (RCC). The referral is triaged based on their age, diagnosis, complexity of care and reason for referral, and is faxed to the appropriate diabetes education program for service. The process ends when the outcome of the initial appointment is known.

CI receives referral from provider → Referral Triaged → Referral sent to DEP by Fax → Information captured in Database → DEP contacts patient to book appointment → DEP records date of first contact with patient and faxes back to CI → CI communicates to referring physician date and location of appointment

Conclusions

- A single point of contact for all patients with diabetes requiring education and for referring health care providers
- Simple and more timely access to information regarding the status of referrals (pending, booked, complete, reported) for primary care providers and potentially patients
- A central contact point for information regarding system availability and where specific types of education interventions are being provided
- Triaging of referrals to provide appropriate priority triaging and distribution of requests to various diabetes education programs
- Better consistency in reporting for accessing capacity
- On-going communication to provide a better source of quality data for system resource planning and best practice sharing

Stand up to Diabetes

Implementation Plan

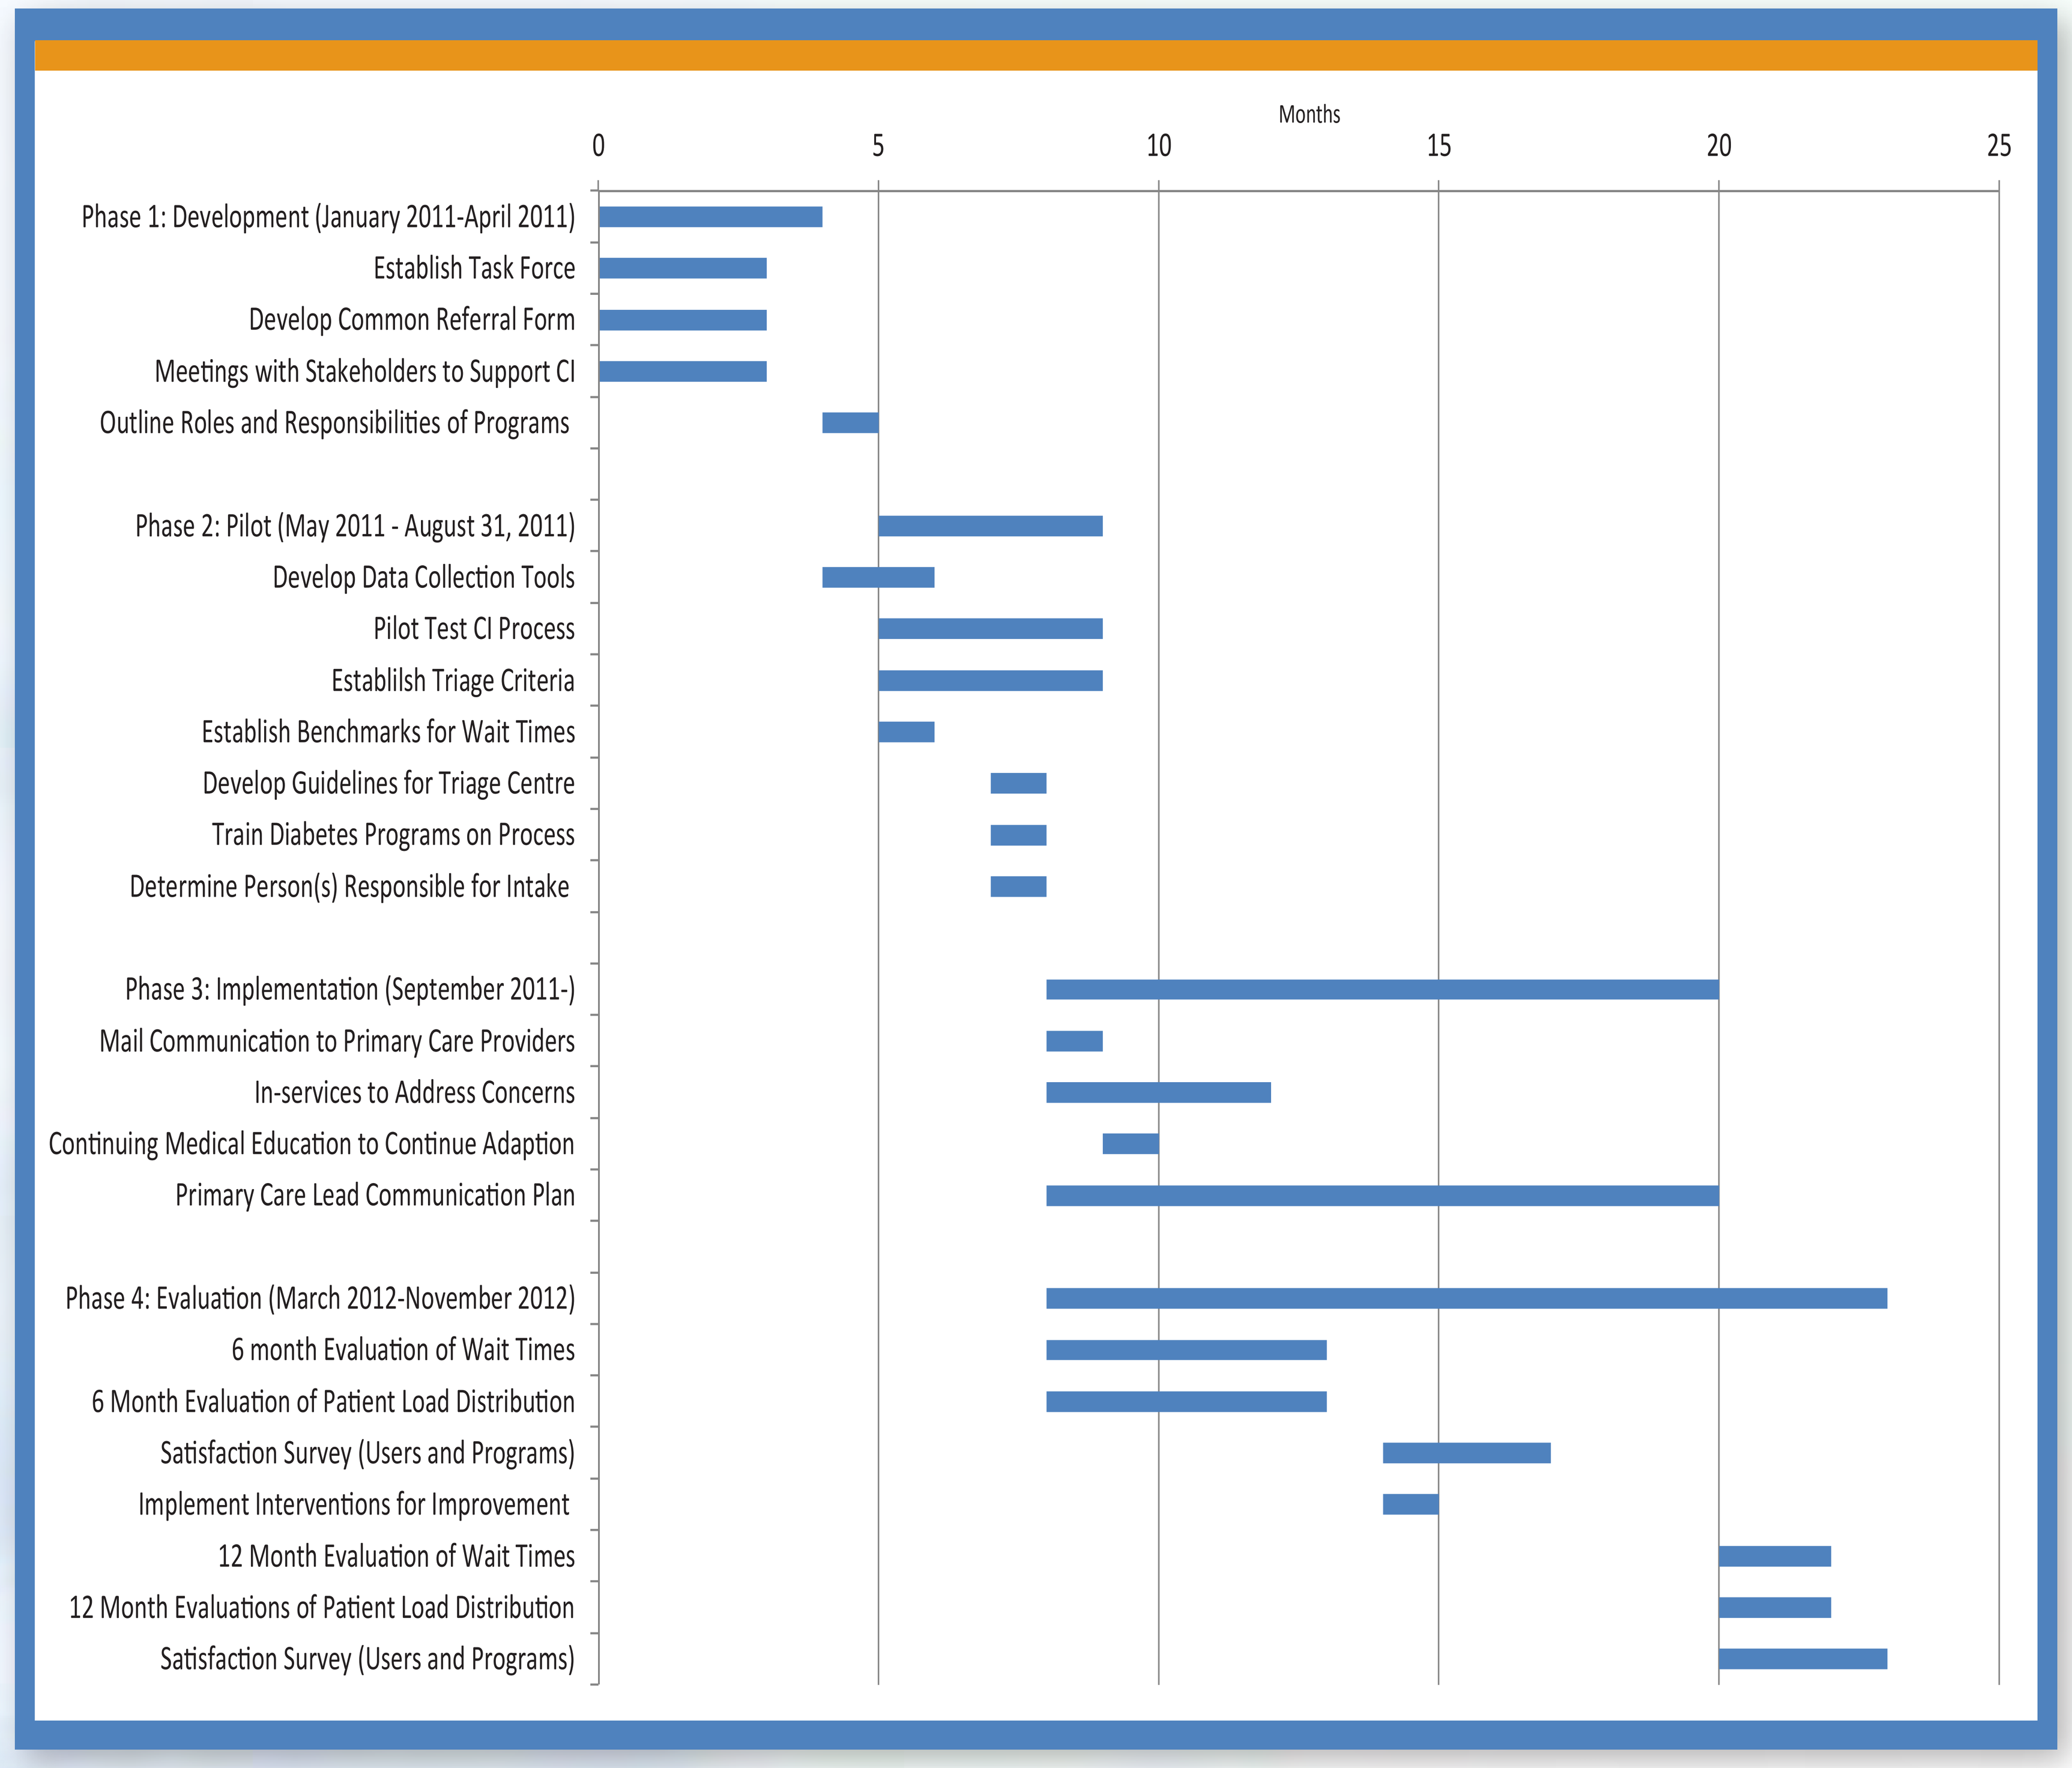
Months
0
5
10
15
20
25
Phase 1: Development (January 2011-April 2011)
Establish Task Force
Develop Common Referral Form
Meetings with Stakeholders to Support CI
Outline Roles and Responsibilities of Programs
Phase 2: Pilot (May 2011 - August 31, 2011)
Develop Data Collection Tools
Pilot Test CI Process
Establilsh Triage Criteria
Establish Benchmarks for Wait Times
Develop Guidelines for Triage Centre
Train Diabetes Programs on Process
Determine Person(s) Responsible for Intake
Phase 3: Implementation (September 2011-)
Mail Communication to Primary Care Providers
In-services to Address Concerns
Continuing Medical Education to Continue Adaption
Primary Care Lead Communication Plan
Phase 4: Evaluation (March 2012-November 2012)
6 month Evaluation of Wait Times
6 Month Evaluation of Patient Load Distribution
Satisfaction Survey (Users and Programs)
Implement Interventions for Improvement
12 Month Evaluation of Wait Times
12 Month Evaluations of Patient Load Distribution
Satisfaction Survey (Users and Programs)

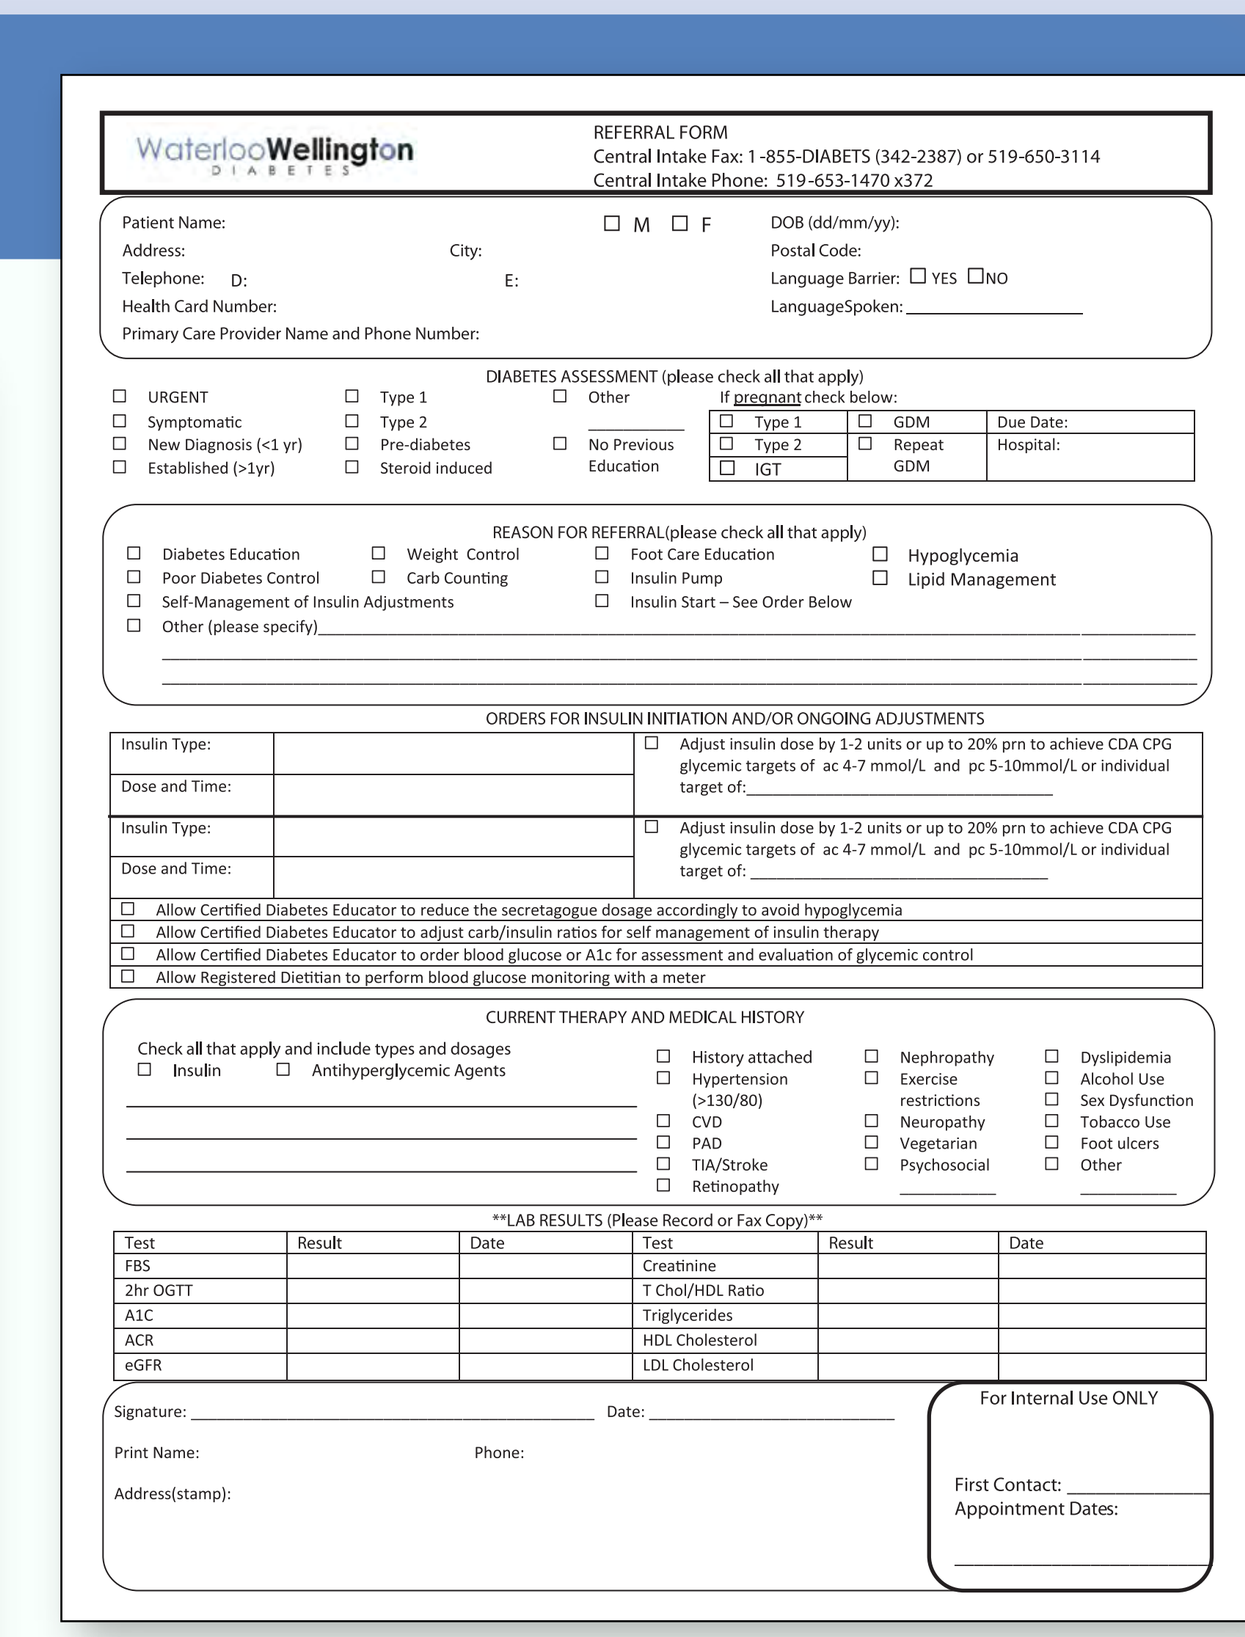

REFERRAL FORM
Central Intake Fax: 1-855-DIABETS (342-2387) or 519-650-3114
Central Intake Phone: 519-653-1470 x372

DIABETES ASSESSMENT (please check all that apply)

REASON FOR REFERRAL (please check all that apply)

ORDERS FOR INSULIN INITIATION AND/OR ONGOING ADJUSTMENTS

CURRENT THERAPY AND MEDICAL HISTORY

LAB RESULTS (Please Record or Fax Copy)

For Internal Use ONLY

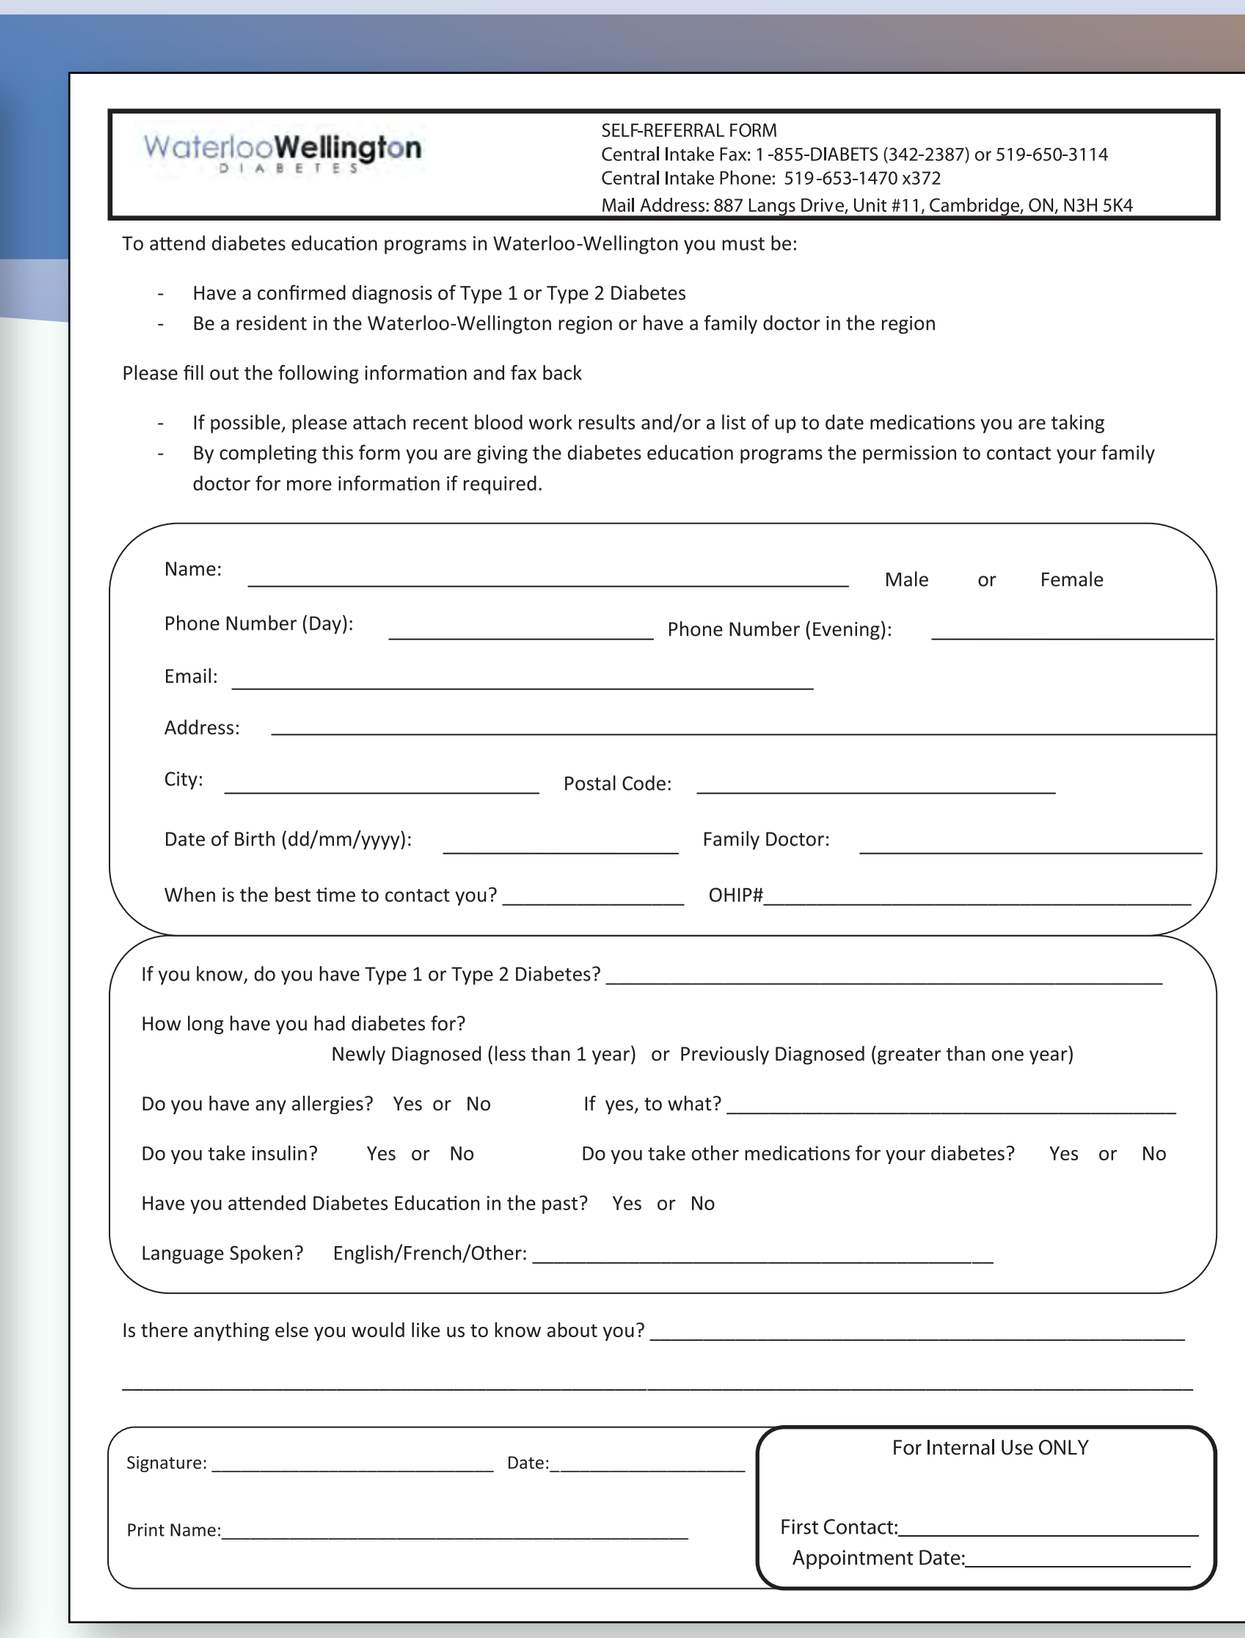

SELF-REFERRAL FORM
Central Intake Fax: 1-855-DIABETS (342-2387) or 519-650-3114
Central Intake Phone: 519-653-1470 x372
Mail Address: 887 Langs Drive, Unit #11, Cambridge, ON, N3H 5K4

To attend diabetes education programs in Waterloo-Wellington you must be:
- Have a confirmed diagnosis of Type 1 or Type 2 Diabetes
- Be a resident in the Waterloo-Wellington region or have a family doctor in the region

Please fill out the following information and fax back
- If possible, please attach recent blood work results and/or a list of up to date medications you are taking
- By completing this form you are giving the diabetes education programs the permission to contact your family doctor for more information if required.

Name: Male or Female
Phone Number (Day): Phone Number (Evening):
Email:
Address:
City: Postal Code:
Date of Birth (dd/mm/yyyy): Family Doctor:
When is the best time to contact you? OHIP#

If you know, do you have Type 1 or Type 2 Diabetes?
How long have you had diabetes for?
Newly Diagnosed (less than 1 year) or Previously Diagnosed (greater than one year)
Do you have any allergies? Yes or No If yes, to what?
Do you take insulin? Yes or No Do you take other medications for your diabetes? Yes or No
Have you attended Diabetes Education in the past? Yes or No
Language Spoken? English/French/Other:

Is there anything else you would like us to know about you?

Signature: Date:
Print Name:

For Internal Use ONLY
First Contact:
Appointment Date:

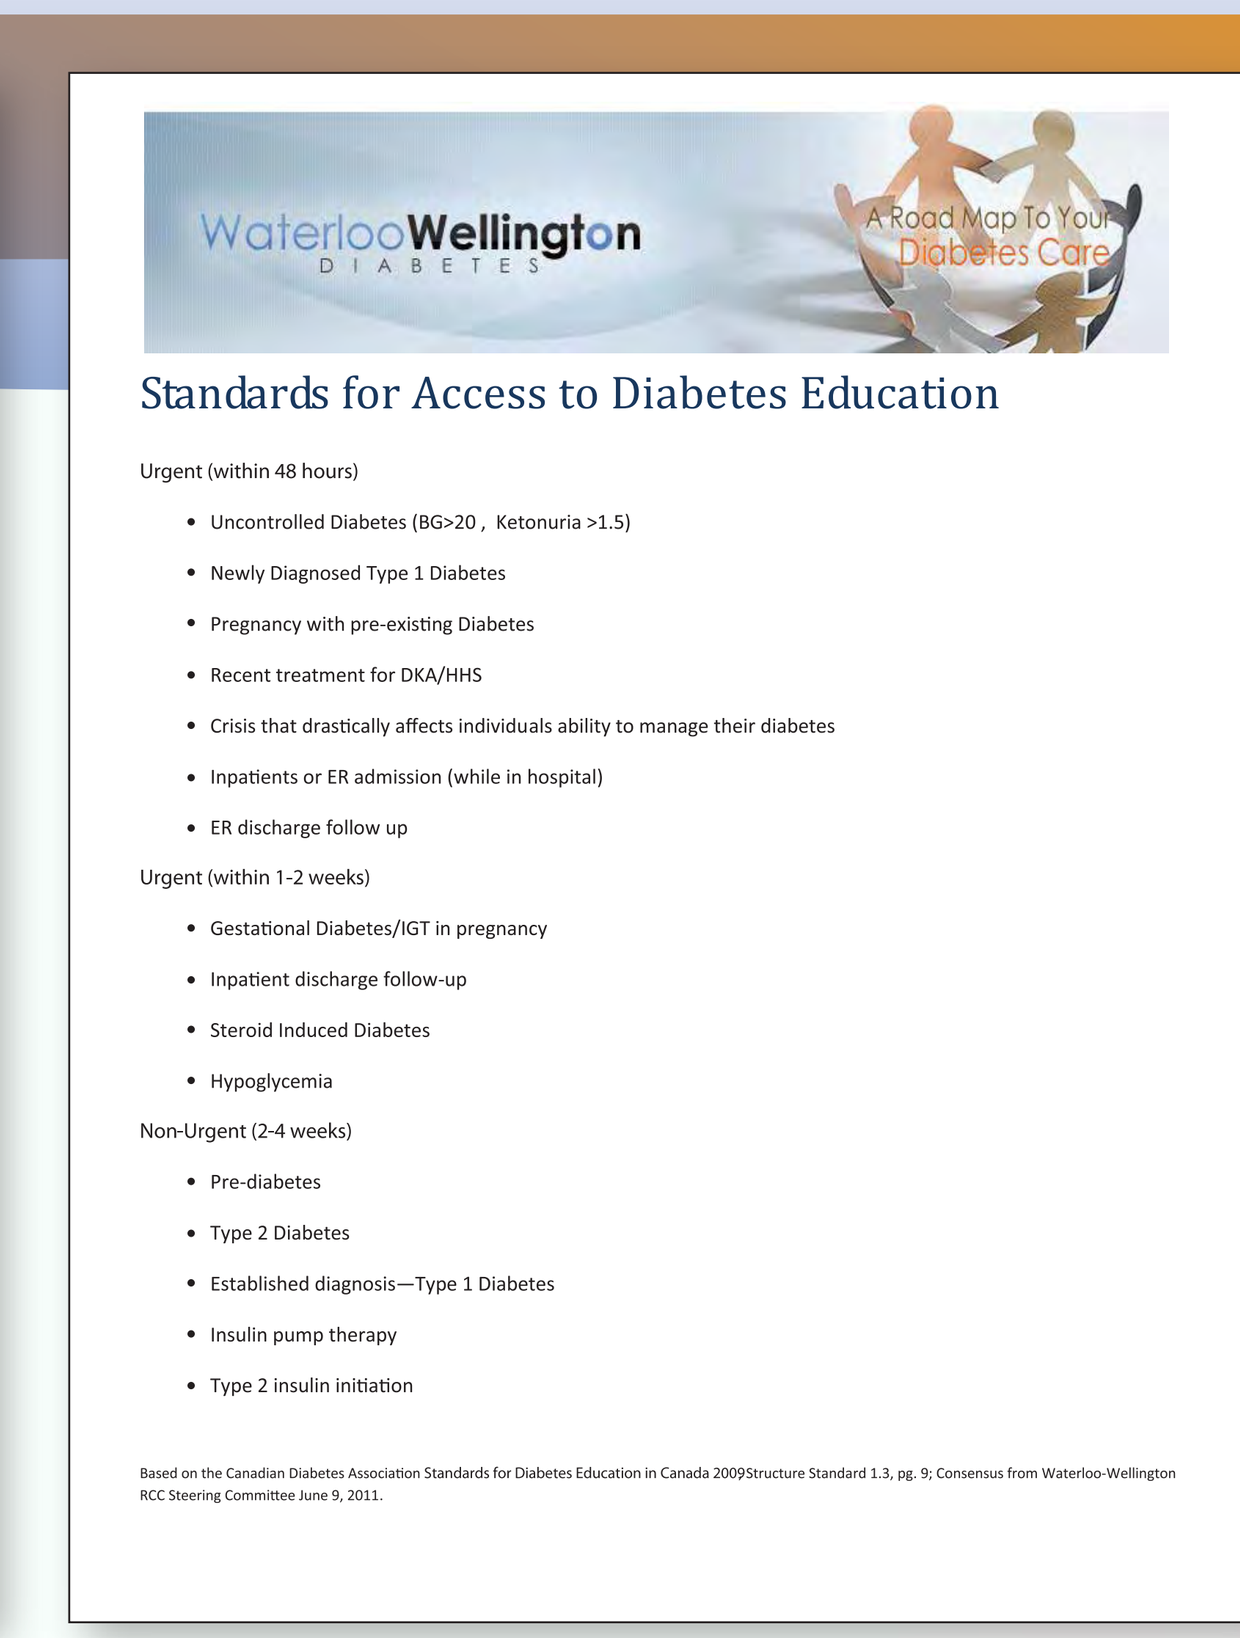

A Road Map To Your Diabetes Care

Standards for Access to Diabetes Education

Urgent (within 48 hours)
- Uncontrolled Diabetes (BG>20, Ketonuria >1.5)
- Newly Diagnosed Type 1 Diabetes
- Pregnancy with pre-existing Diabetes
- Recent treatment for DKA/HHS
- Crisis that drastically affects individuals ability to manage their diabetes
- Inpatients or ER admission (while in hospital)
- ER discharge follow up

Urgent (within 1-2 weeks)
- Gestational Diabetes/IGT in pregnancy
- Inpatient discharge follow-up
- Steroid Induced Diabetes
- Hypoglycemia

Non-Urgent (2-4 weeks)
- Pre-diabetes
- Type 2 Diabetes
- Established diagnosis—Type 1 Diabetes
- Insulin pump therapy
- Type 2 insulin initiation

Based on the Canadian Diabetes Association Standards for Diabetes Education in Canada 2009Structure Standard 1.3, pg. 9; Consensus from Waterloo-Wellington RCC Steering Committee June 9, 2011.

Diabetes Care Continuum Roles and Responsibilities

Diabetes Clinic Visit
- Primary care provider quarterly visit
- May involve support staff i.e. RN/RD
- Assess patient's control and management of chronic disease
- Utilize patient flow sheet as guide sheet
- Management of medications to control blood glucose levels

Diabetes Education Programs
- Diabetes Programs provide basic to intermediate level diabetes education and management services through a model that is needs-based and community-based
- Type 2 Diabetes New Diagnosis and Established
- High Risk Pre-Diabetes Populations
- Specialized Classes
 - Diabetes A, B, C
 - Hypertension Management
 - Lipid Management
 - Foot Care Education
 - Meter Teaching
- Urgent Insulin Starts
- Insulin Management

Complex Diabetes Care Centres
- Complex Diabetes Care Centres provide intermediate to advanced level diabetes education and management services through a model that is needs-based and community-based
- Complex Insulin Management
 - Multiple Daily Injections
 - Carbohydrate Counting
- Hypoglycemia Unawareness
- Urgent Insulin starts
- Insulin Pump Therapy
- Gestational Diabetes Management
- Pre-conceptual counselling
- Pregnant Type 1 and Type 2
- Inpatient Care/Emergency Care Pathway
- Steroid-Induced Diabetes
- Type 1 Adult & Paediatrics
- Transitional Programming
- Continuous Glucose Monitoring
- Partnership with Renal Programs
- Specialized Classes

All Patients (New and Established) Needing Education and Support →

← Well Controlled Patients

↔ Patients unable to meet targets within recommended timeframe who need to be reassessed

Results

Physician and DEP Satisfaction Surveys: There was consensus amongst all physicians and DEP staff that they have been waiting for a simple process with one form for some time and they were very satisfied with the process. An additional important element in the referring physician's satisfaction is the receipt of information on the patient appointment within a reasonable period.

Quantitative Data Analysis: During a twelve month period, a total of 1917 referrals were received. On average, CI processes about 160 referrals monthly. The distribution of these referrals is displayed below

Number of Referrals Received at Central Intake, June 01, 2011 – May 31, 2012

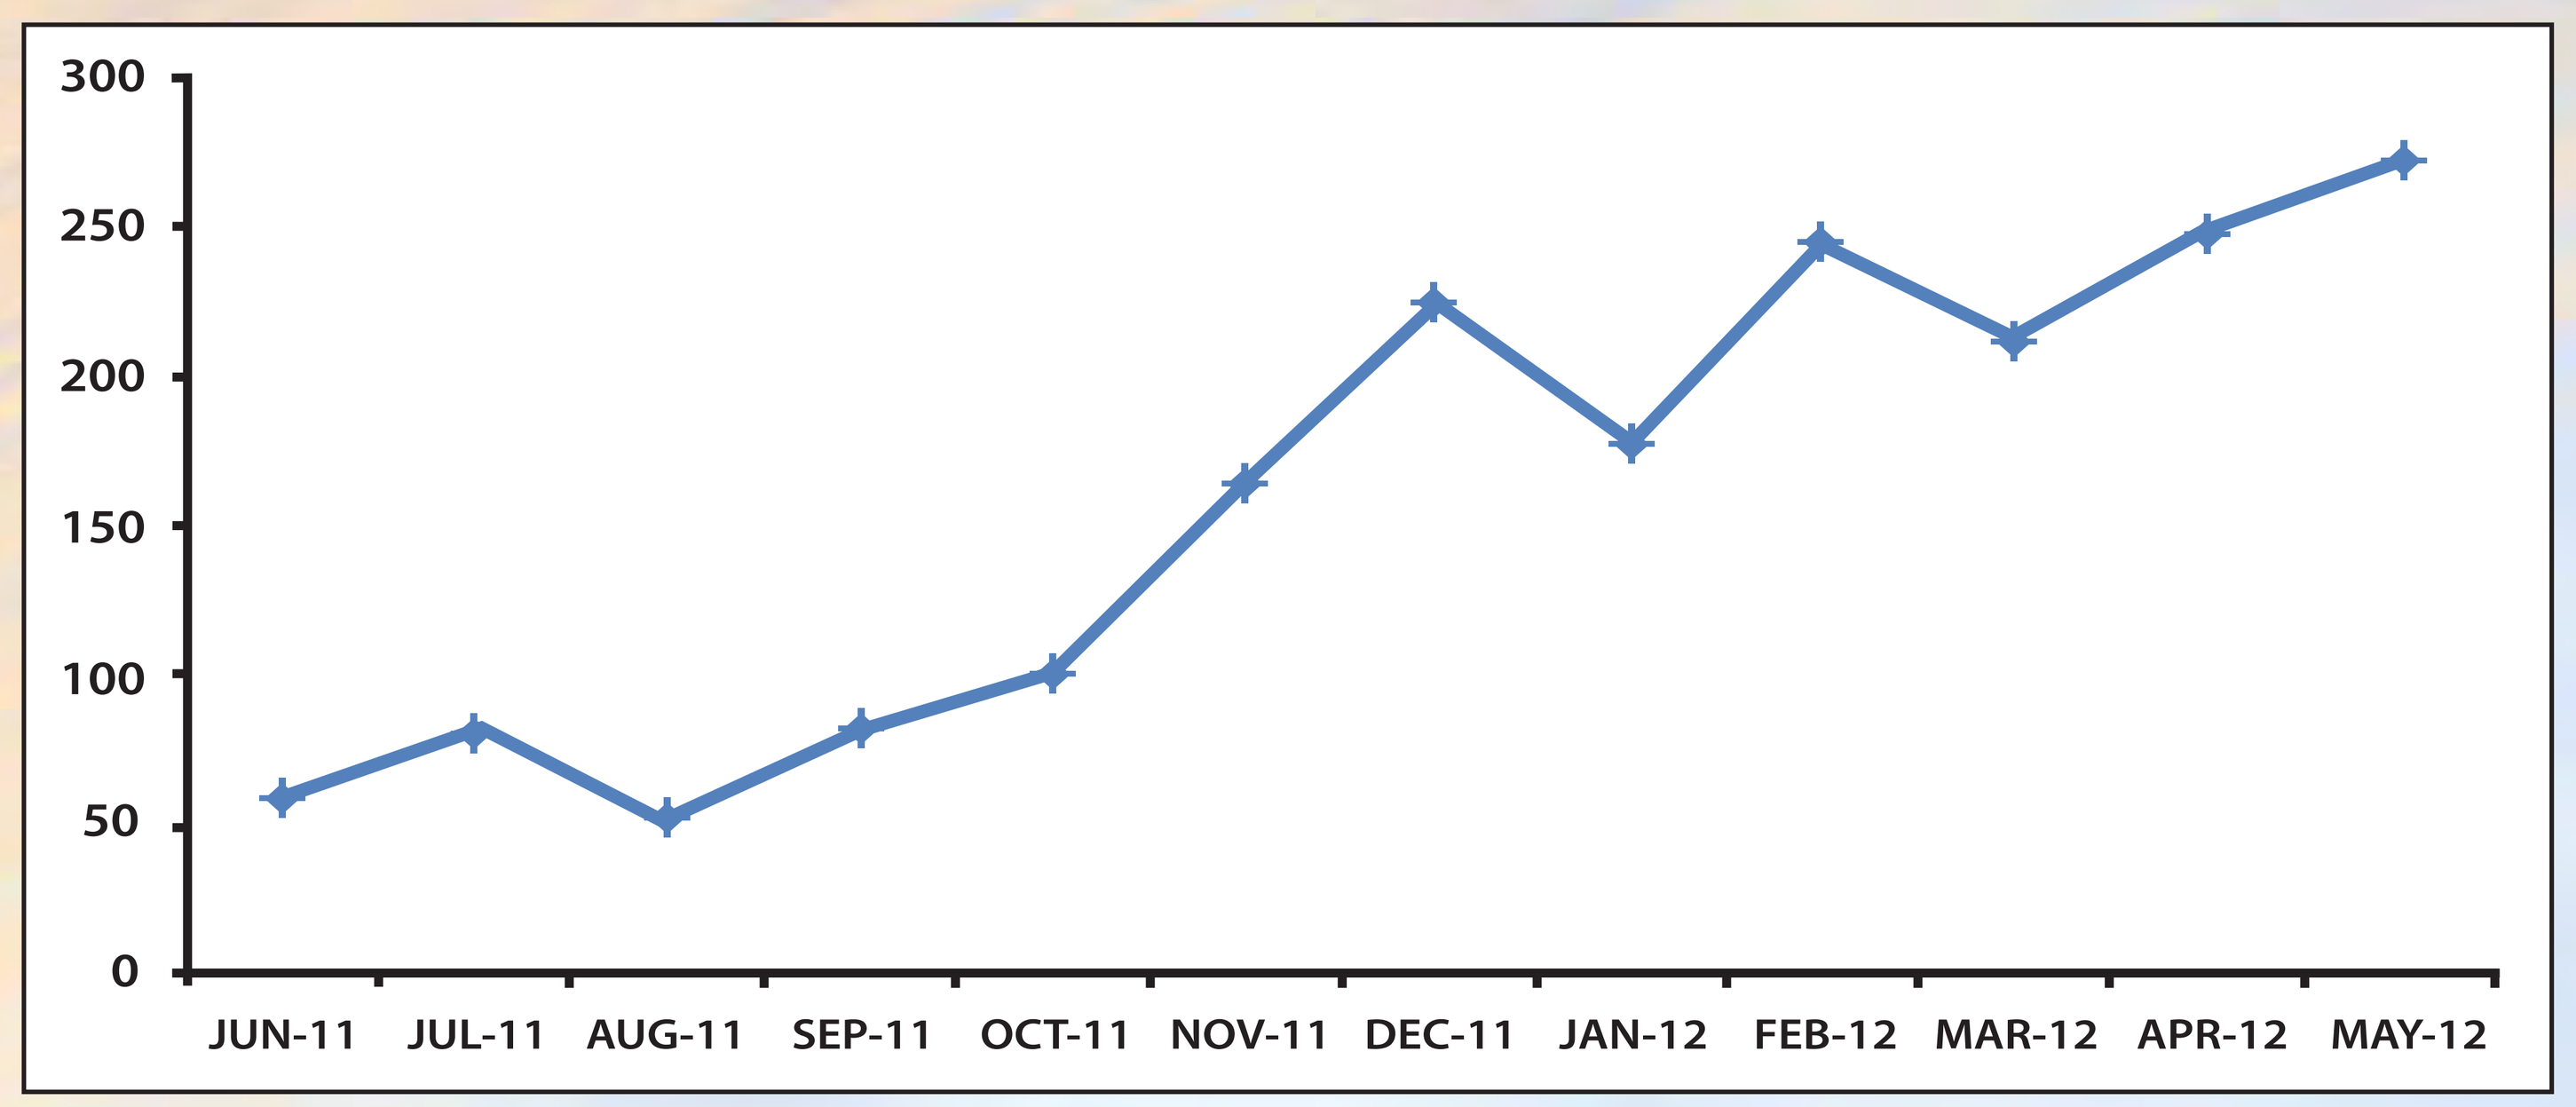
300
250
200
150
100
50
0
JUN-11
JUL-11
AUG-11
SEP-11
OCT-11
NOV-11
DEC-11
JAN-12
FEB-12
MAR-12
APR-12
MAY-12

Number of Referrals with Established and Newly Diagnosed Diabetes, June 01, 2011 – May 31, 2012

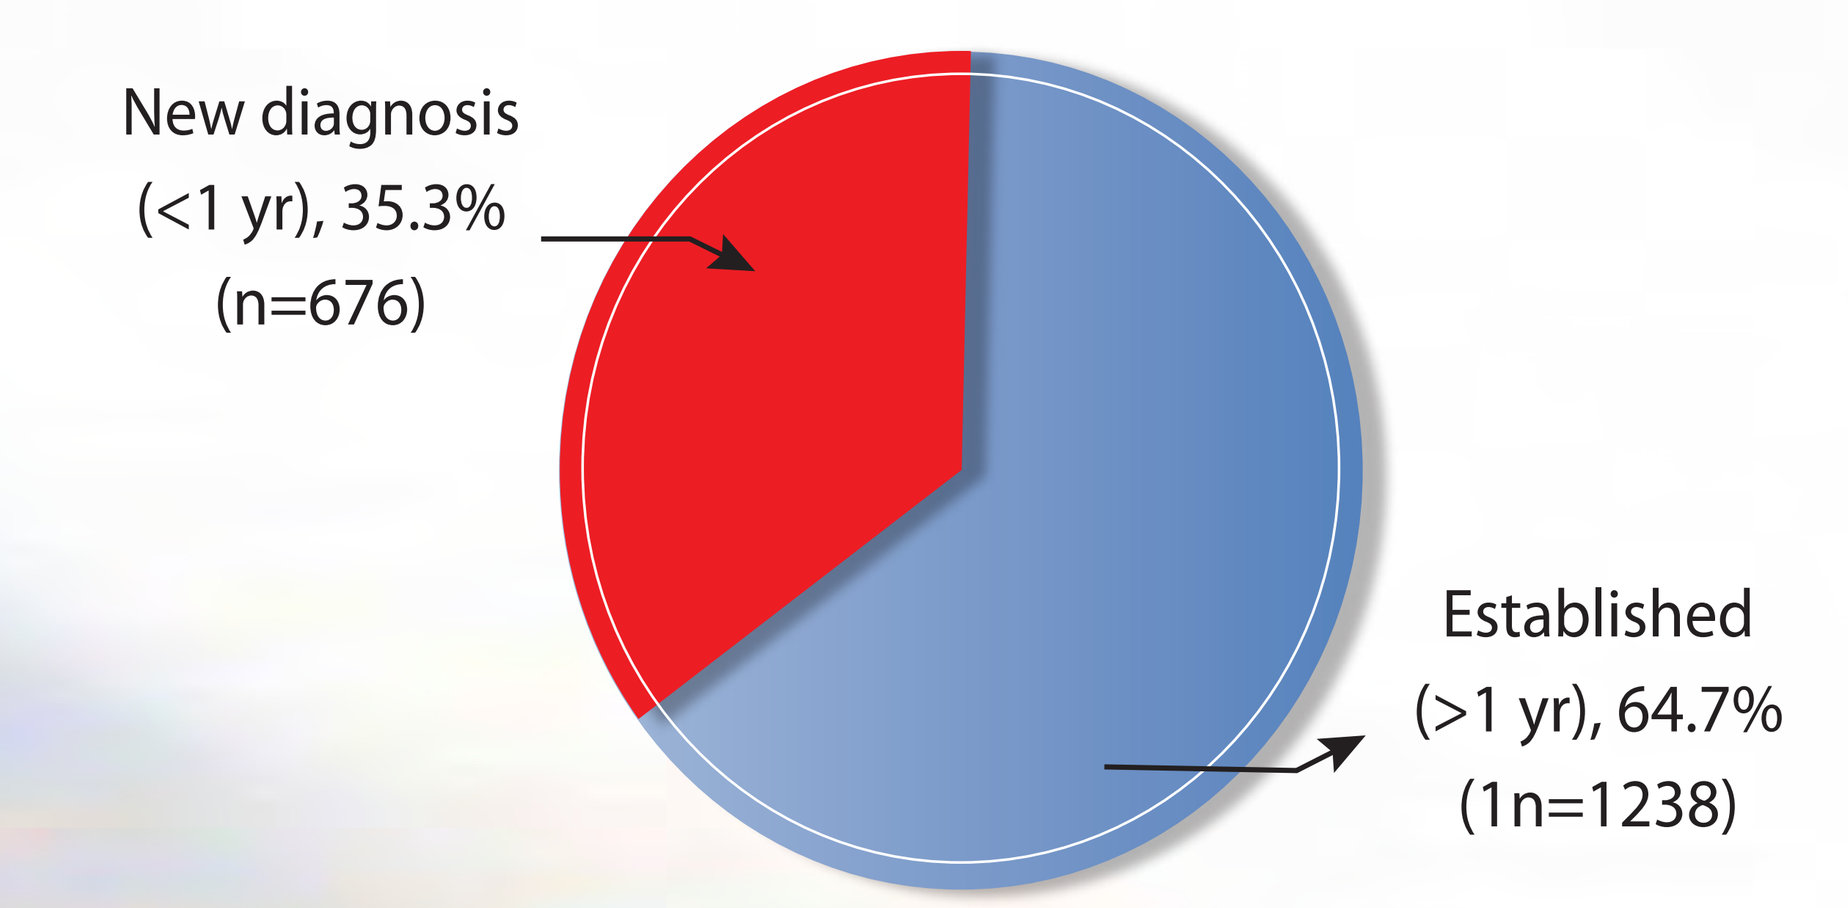
New diagnosis
(<1 yr), 35.3%
(n=676)
Established
(>1 yr), 64.7%
(1n=1238)

Patterns of Referral according to Specialty, June 01, 2011 - May 31, 2012

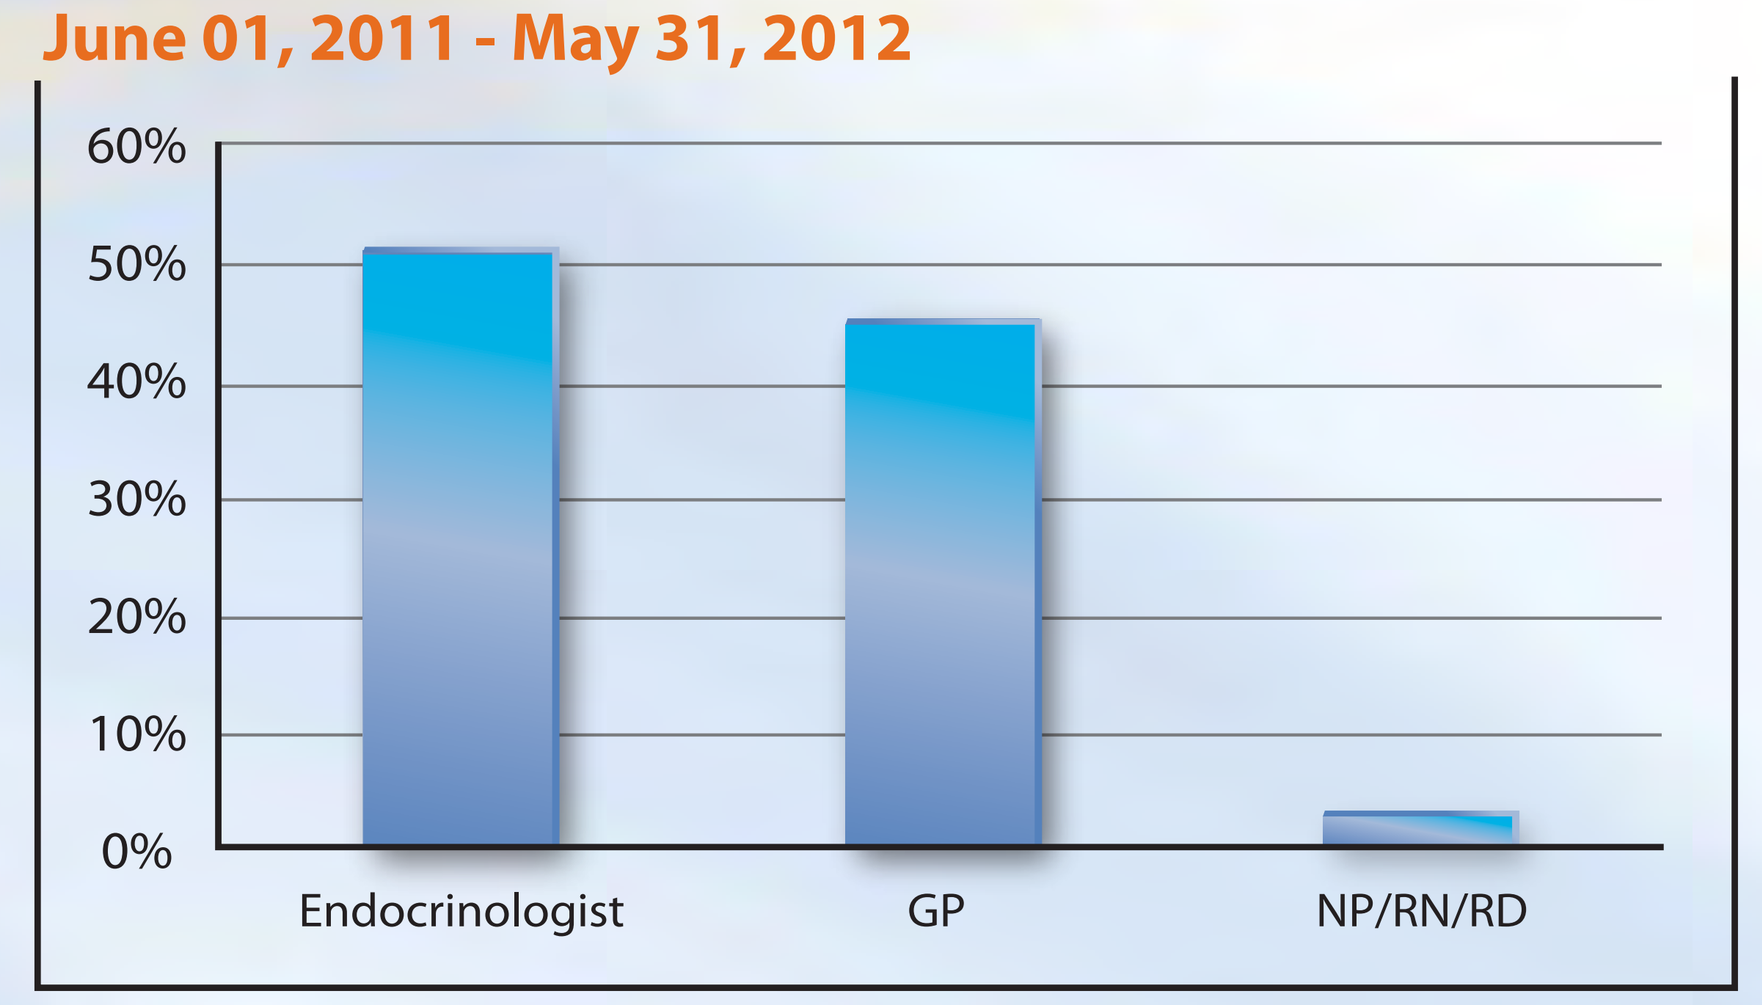
60%
50%
40%
30%
20%
10%
0%
Endocrinologist
GP
NP/RN/RD

Wait Times (in days) by Diabetes Type **June 1, 2011 - May 11, 2012**

Diabetes Type	No. referrals through CI	No. booked appt. with educator	% booked appt. with educator	SUM Wait Times (days)	Mean Wait Times (days)
Type 2	1247	1048	84.0%	32233	31
Type 1	316	259	82.0%	9599	37
Pre-diabetes	198	185	93.4%	8163	44
GDM	71	50	70.4%	397	8
IGT of Pregnancy	20	19	95.0%	93	5
Pregnant Type 2	19	17	89.5%	168	10
Repeat GDM	13	11	84.6%	68	6
Pregnant Type 1	12	11	91.7%	158	14
Other	9	8	88.9%	316	40
Steroid induced	<6	<6	S	S	S
Total	1910	1613	84.5%	51264	32

S: data suppressed due to cell size <6.

Community. Health. Wellness.